Effect of Approved Non-Oncology Drugs on the Metastatic Process: Risks & Benefits

EACR 2023 — 12-15 June 2023, Torino, Italy

Debabani Roy Chowdhury[1], Chinmaya Amarkanth[1], Juhi Tayal[2], Ankesh Khemani[1#], Tanvi Singh[1#], Shashank Rao[1#], Sundarajan Kannan[1], Ayushi Gandhi[1] Anurag Mehta[2], Dinesh Doval[2] , John Ellingboe[1] & Arnab Roy Chowdhury[1],

[1]Mestastop Solutions, Bangalore, India & Mestastop Inc, NJ, USA, [2]Rajiv Gandhi Cancer Centre and Research Insitute, New Delhi, India, [#]Advisors – Data Strategy and AI

EACR23-0346

Background

- Most drugs approved for metastatic cancer either treat the secondary tumour's proliferation or influence immunity.
- None of them delays the process of metastasis or has any effect on the pathophysiology of metastasis[1].
- As a result, overall and progression-free survival of solid tumour patients have improved only by 2.8 and 3.3 months over the last decade[2].
- Therefore, It is imperative to identify drugs that would effectively delay metastasis and be relatively safe to be positioned for neo-adjuvant and adjuvant clinical settings.
- As 90% of cancer deaths are due to metastasis, targeting metastasis in combination with other treatments, should be beneficial in improving the low clinical benefit of existing therapies[3].
- Mestastop took a systems biology-based approach and created three proprietary platforms to identify and progress anti-metastatic compounds.
 1. METAssay® - in vitro, dissects metastasis biology[4]
 2. METSCAN® - ex vivo translation of patient samples[5]
 3. METVivo® - In vivo high throughput animal model[6]

Methods

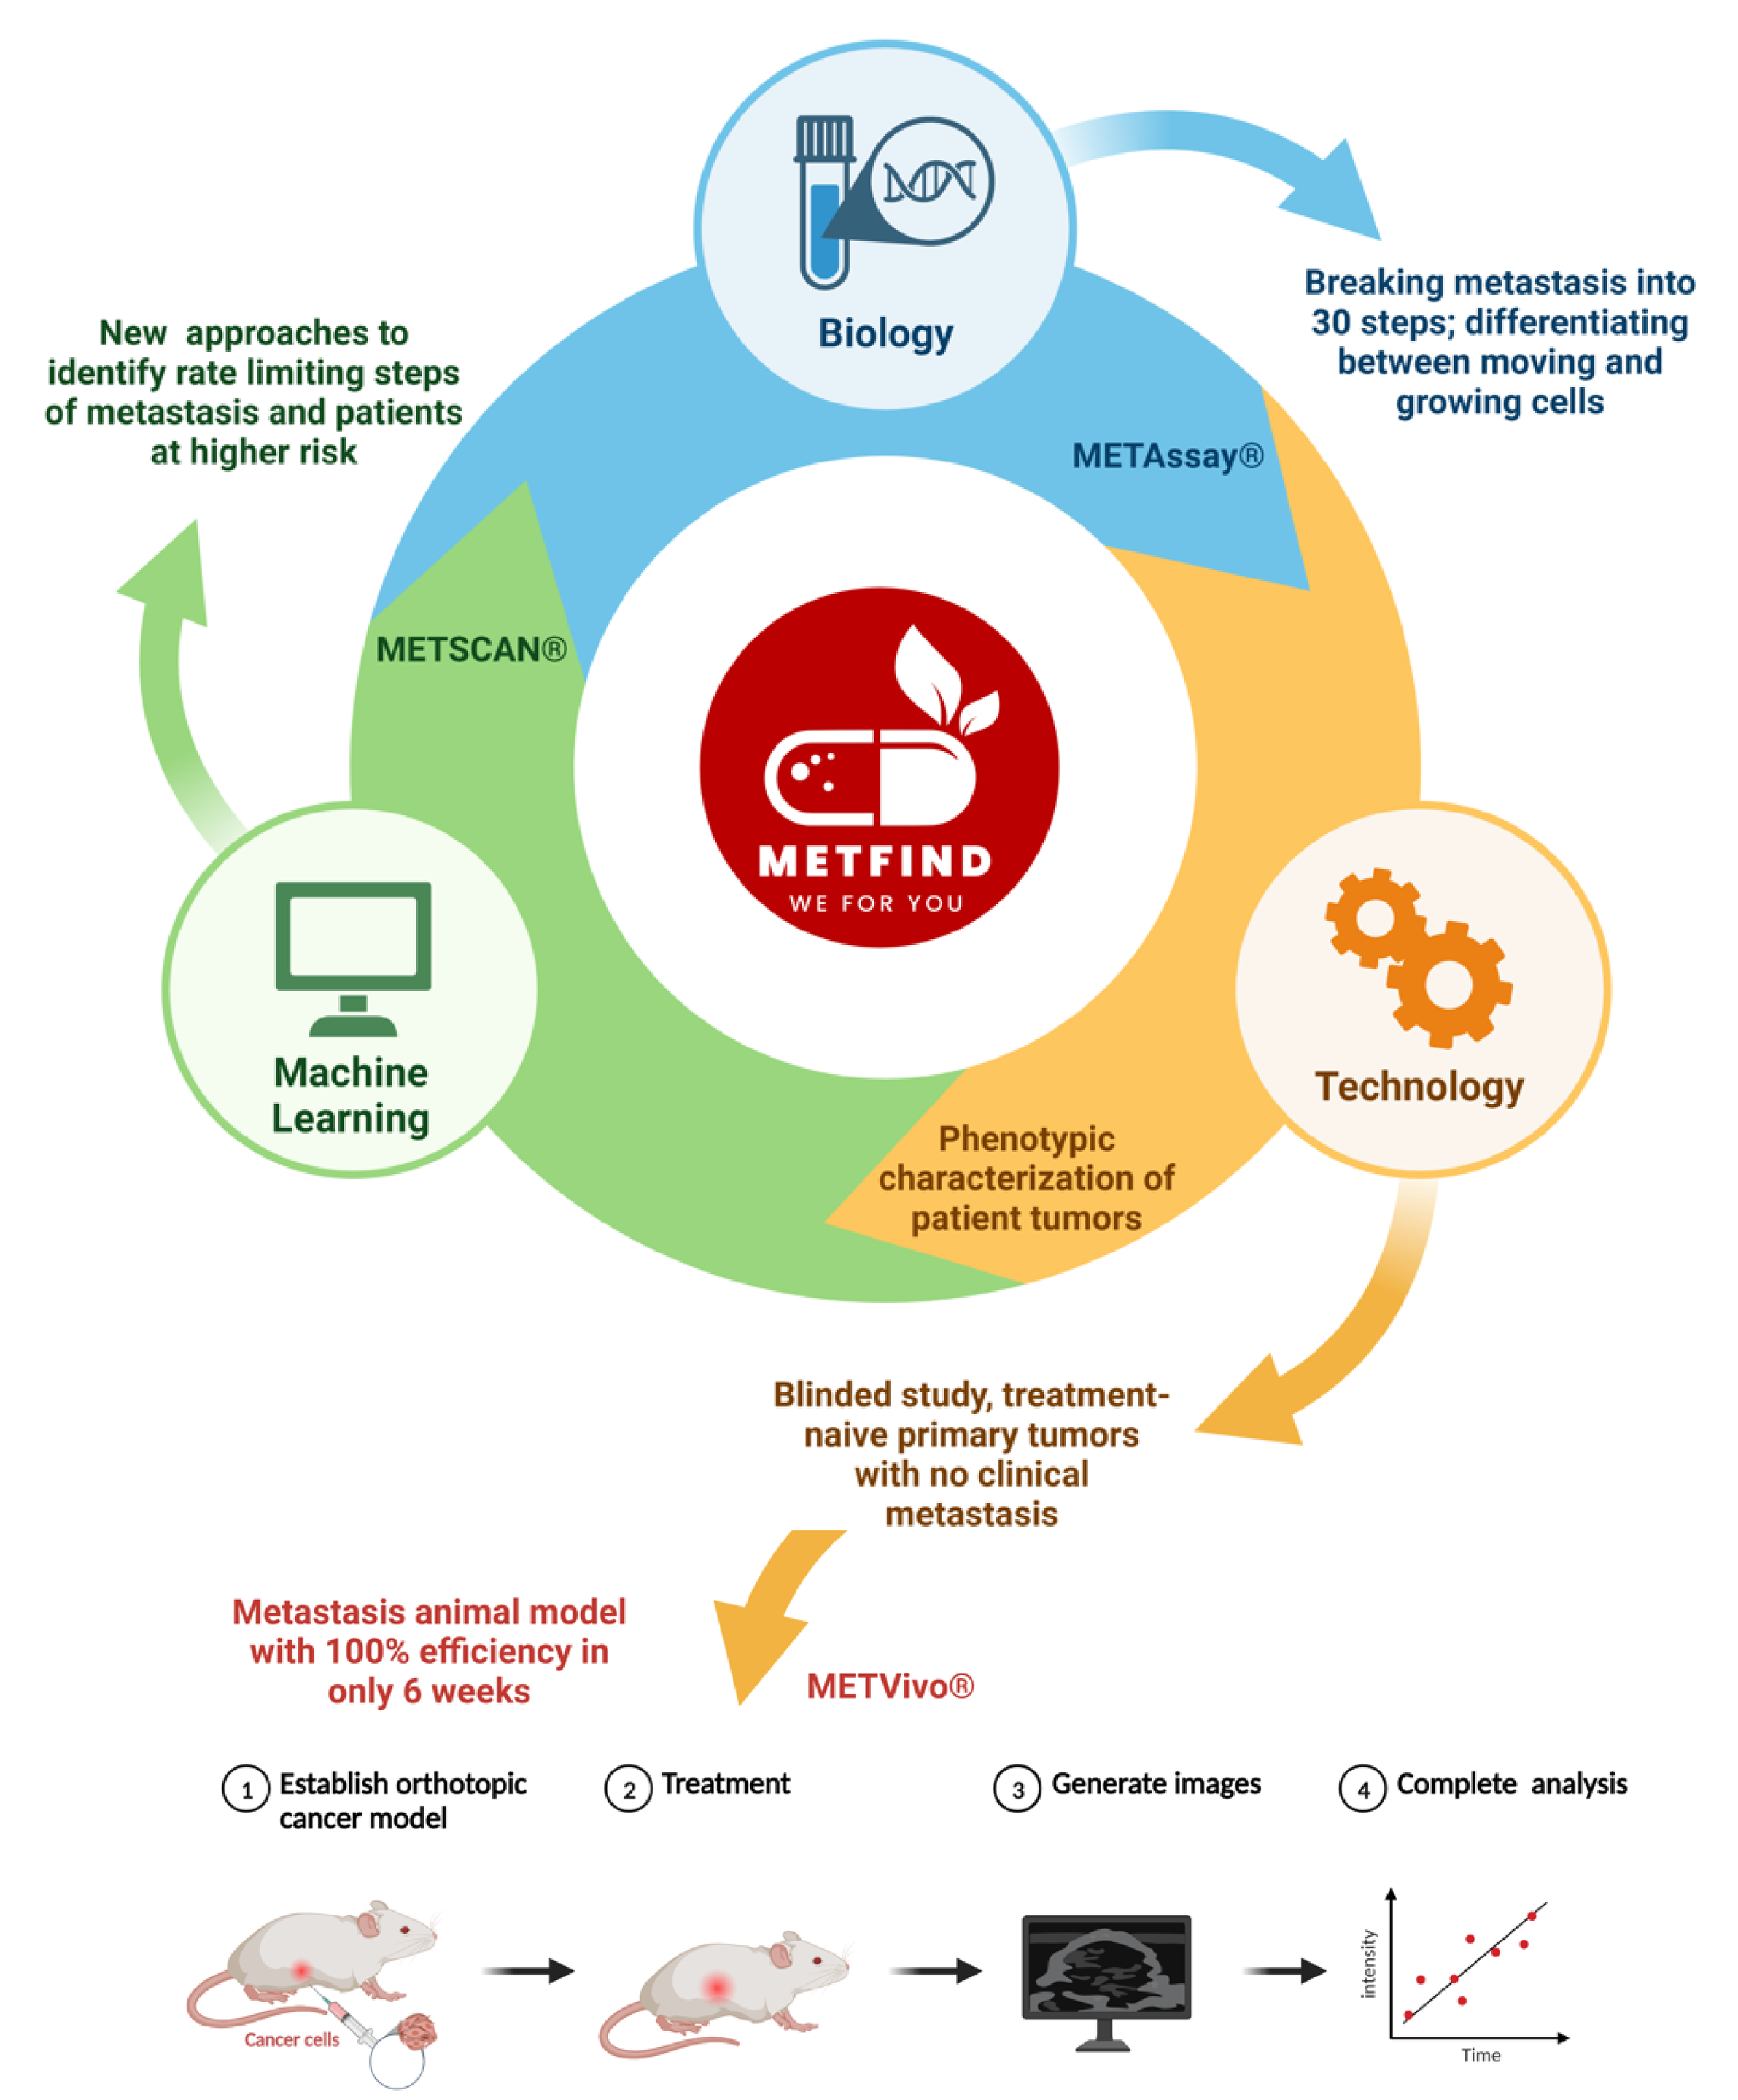
Biology
Breaking metastasis into 30 steps; differentiating between moving and growing cells
METAssay®
New approaches to identify rate limiting steps of metastasis and patients at higher risk
METSCAN®
METFIND
WE FOR YOU
Machine Learning
Technology
Phenotypic characterization of patient tumors
Blinded study, treatment-naive primary tumors with no clinical metastasis
Metastasis animal model with 100% efficiency in only 6 weeks
METVivo®
1 Establish orthotopic cancer model
2 Treatment
3 Generate images
4 Complete analysis
Cancer cells
Time

Fig. 1: Schematics of the Mestastop Platforms

Results — Platform Standardization

Fig.2: (A) METAssay® dissects metastasis biology and (B) differentiates between functional properties of growing and moving cells

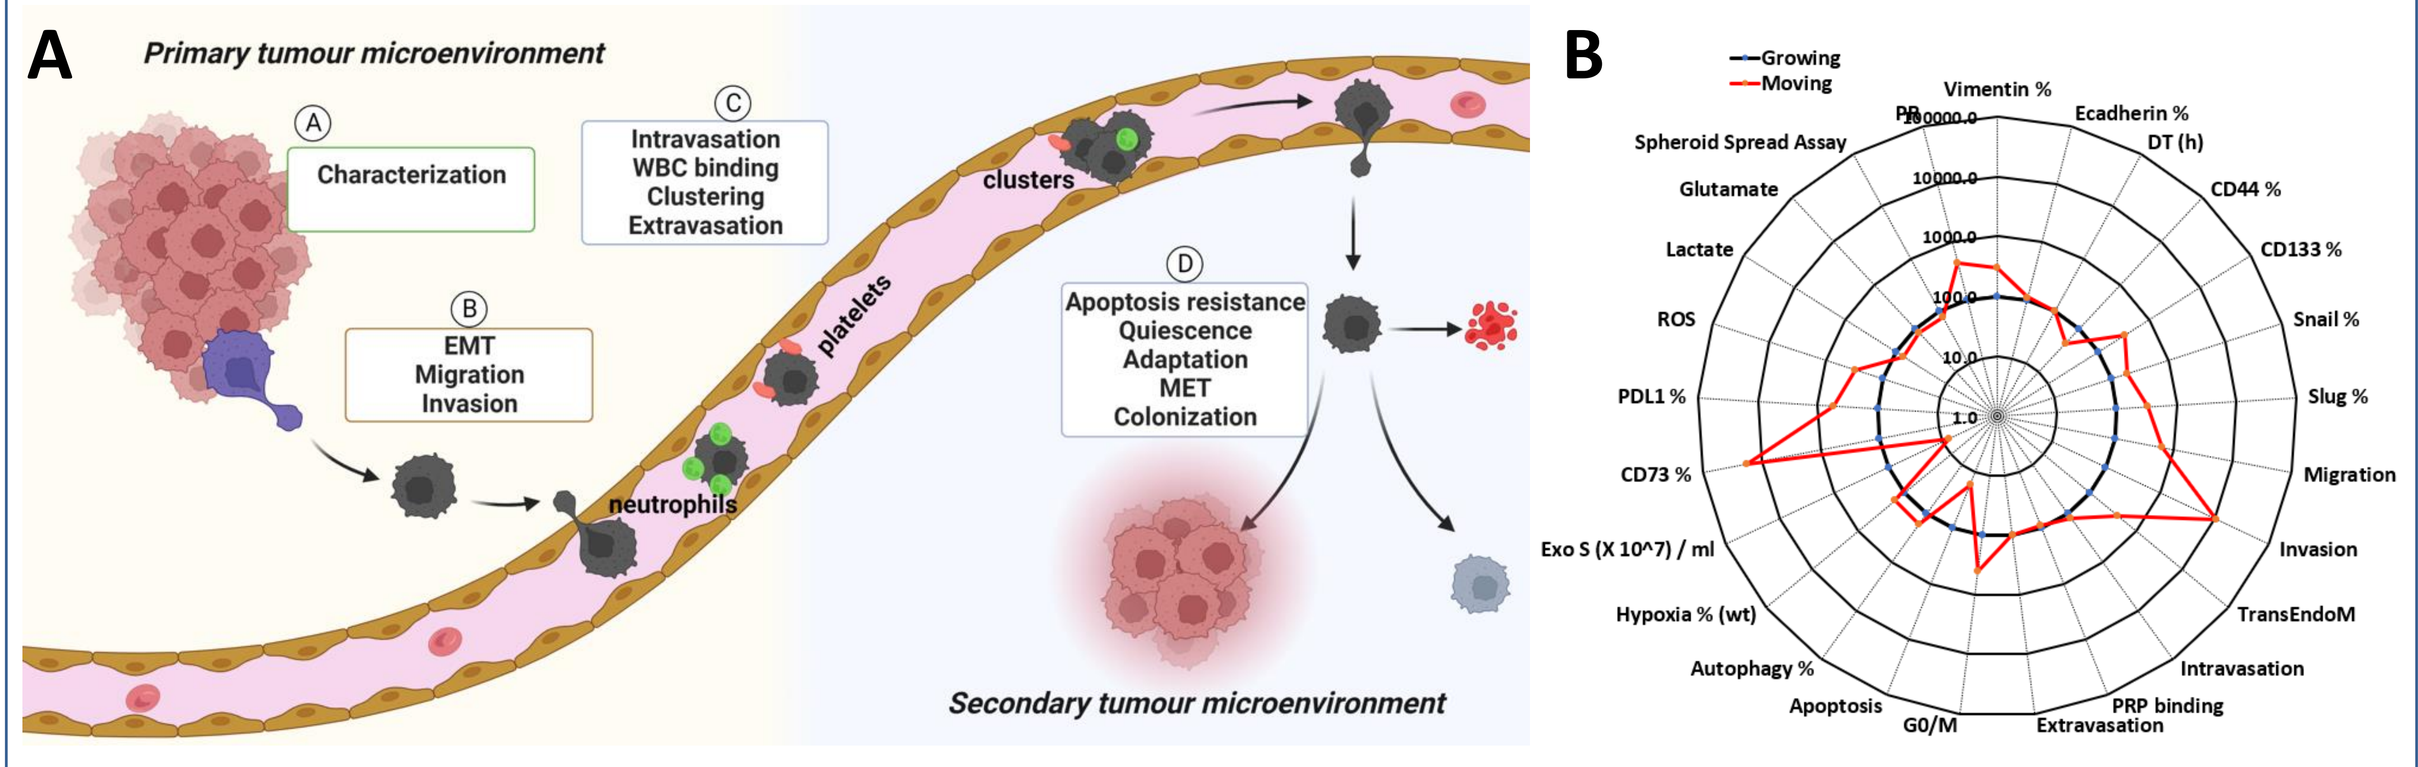
A
Primary tumour microenvironment
Characterization
EMT
Migration
Invasion
Intravasation
WBC binding
Clustering
Extravasation
Apoptosis resistance
Quiescence
Adaptation
MET
Colonization
Secondary tumour microenvironment
B
Growing
Moving

Fig.2: Patients from different tumour types show a similar pattern, (C) CRC & (D) HNC

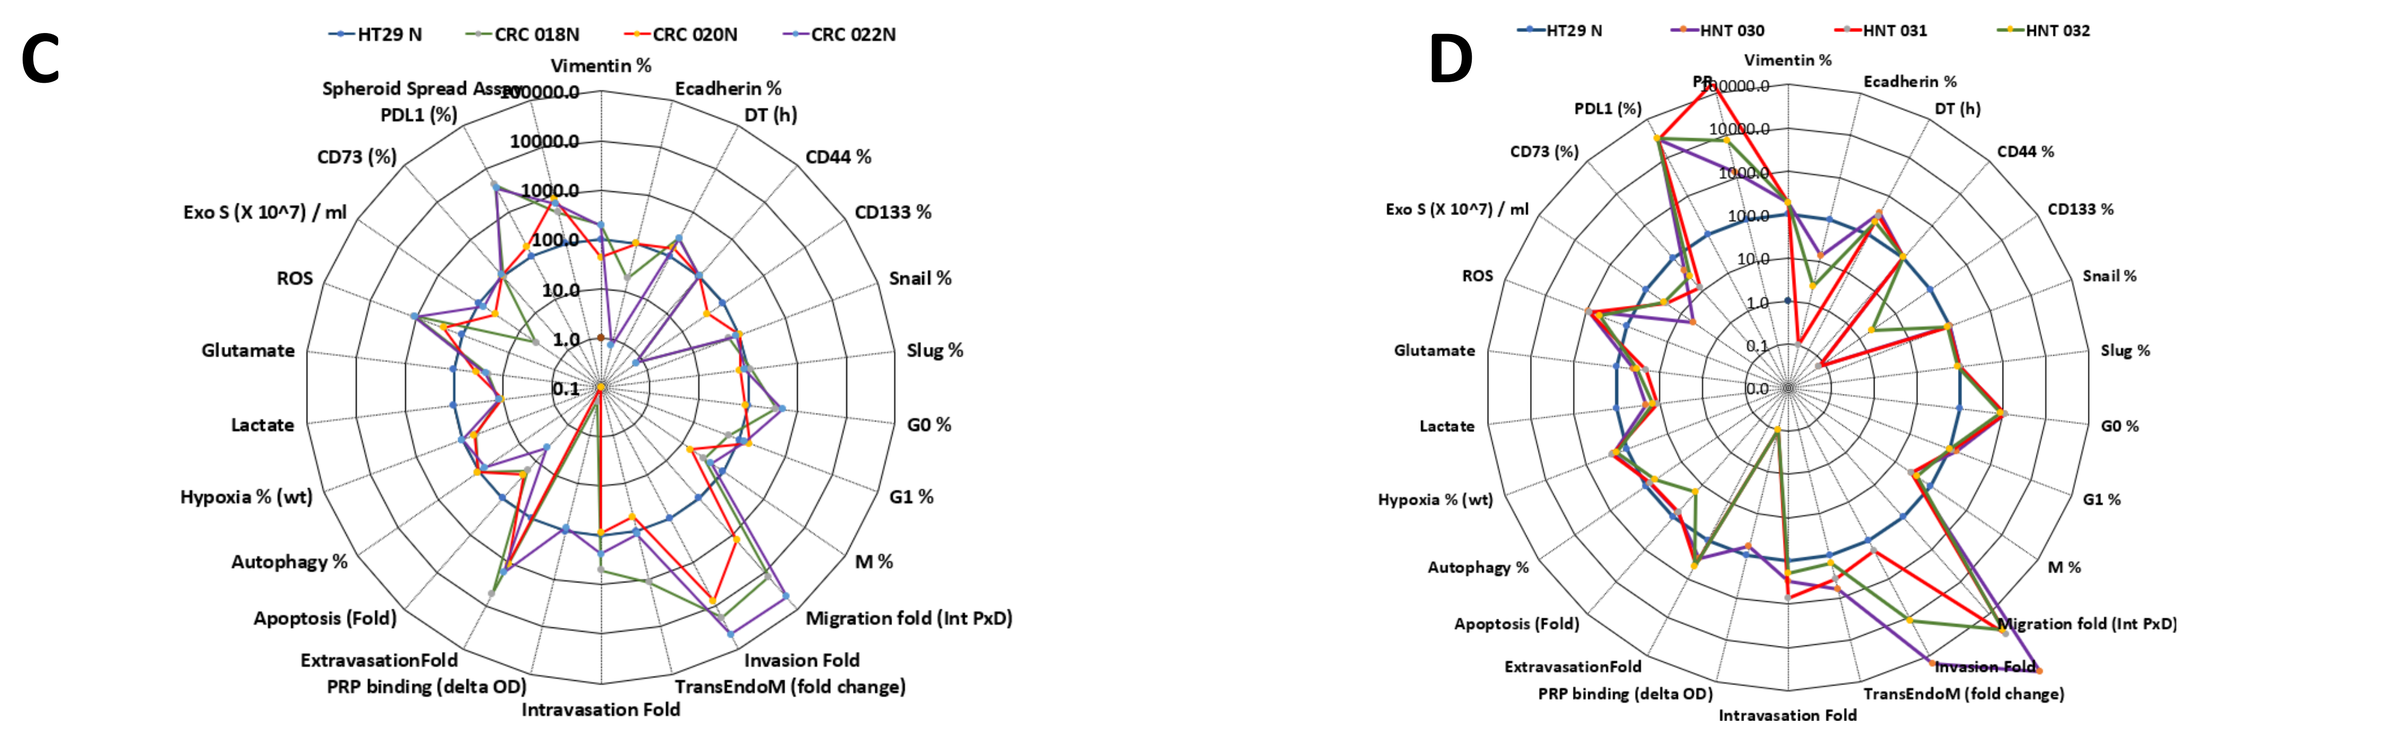

Fig.2: High throughput animal model with (E) low tumorigenicity, (F) efficient metastasis

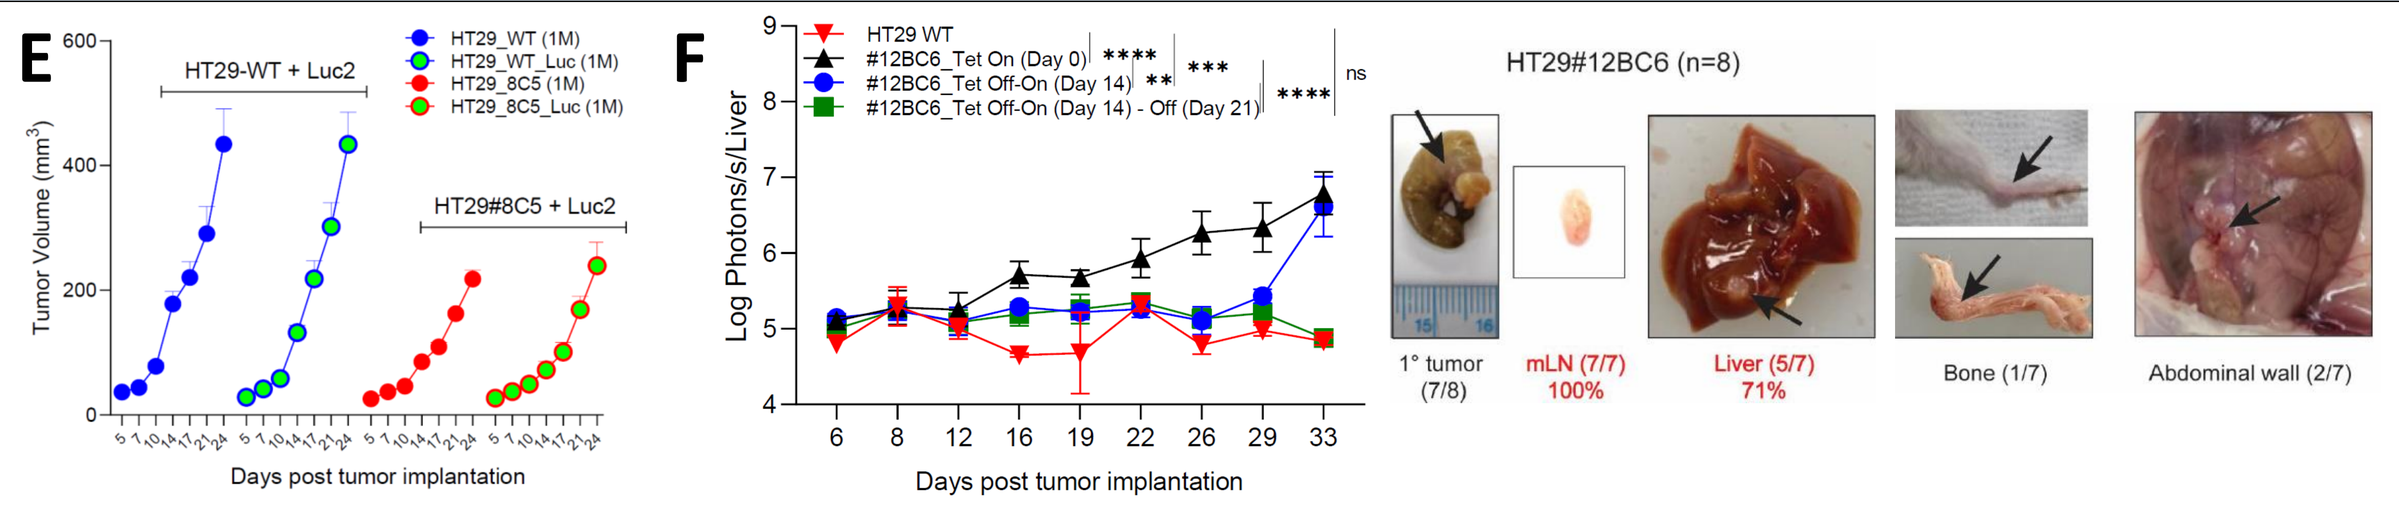
E
HT29-WT + Luc2
HT29#8C5 + Luc2
Days post tumor implantation
F
Log Photons/s/Liver
HT29#12BC6 (n=8)
1° tumor (7/8)
mLN (7/7) 100%
Liver (5/7) 71%
Bone (1/7)
Abdominal wall (2/7)

Result — *In-vitro* to *in-vivo* translation PoC

Fig.3: (A) List of ten approved drugs and their (B) Heat map on key steps of metastasis

A

Code Name	Indications
MS-AP-001	Pain, fever, inflammation
MS-AP-002	Heartburn, peptic ulcer
MS-AP-003	Allergy
MS-AP-004	Hypertension
MS-AP-005	Type II Diabetes
MS-AP-006	Asthma, Allergic Rhinitis
MS-AP-007	Schizophrenia
MS-AP-008	Hypertension
MS-AP-009	Metastatic colon cancer
MS-AP-010	Hypertension

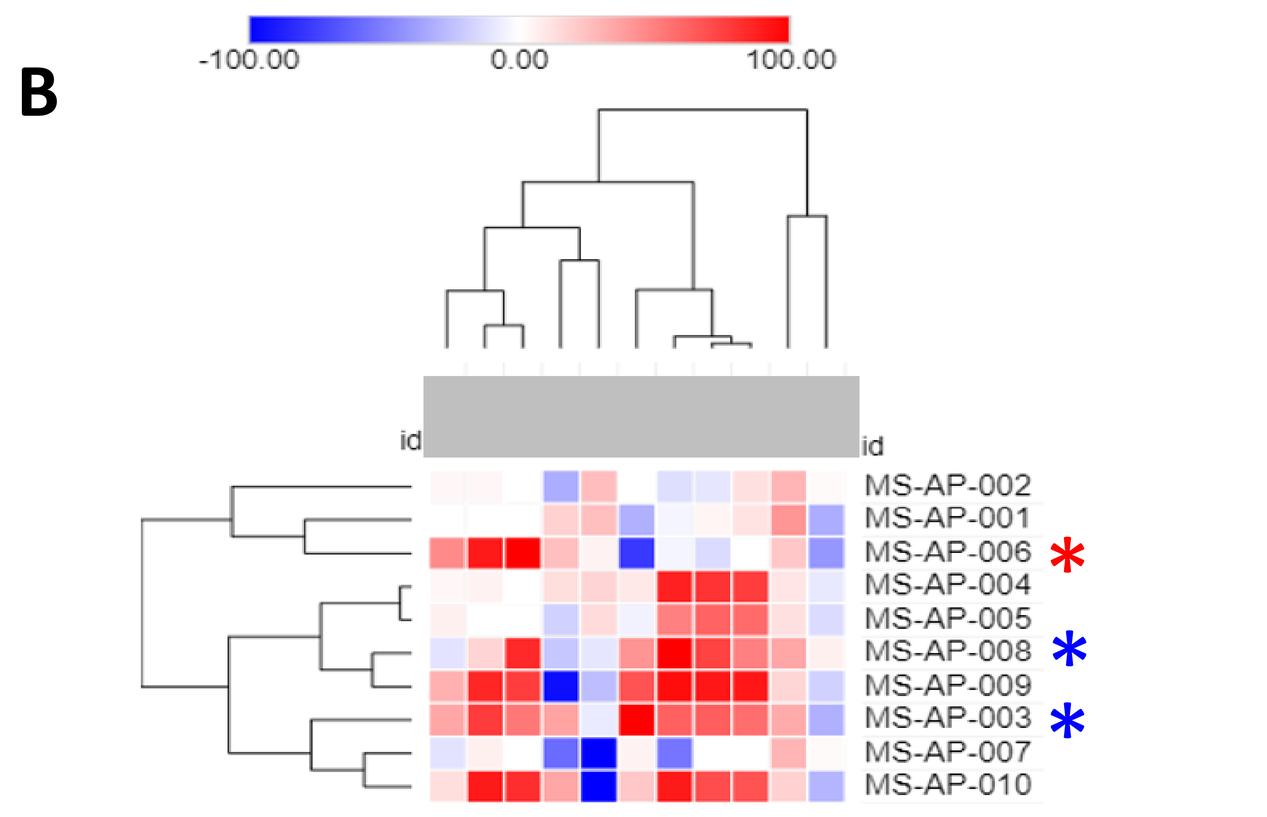
B
-100.00 0.00 100.00
MS-AP-002
MS-AP-001
MS-AP-006
MS-AP-004
MS-AP-005
MS-AP-008
MS-AP-009
MS-AP-003
MS-AP-007
MS-AP-010

Fig.3: Effect of positive compound AP-003 on (C) patients and (D) in animal model

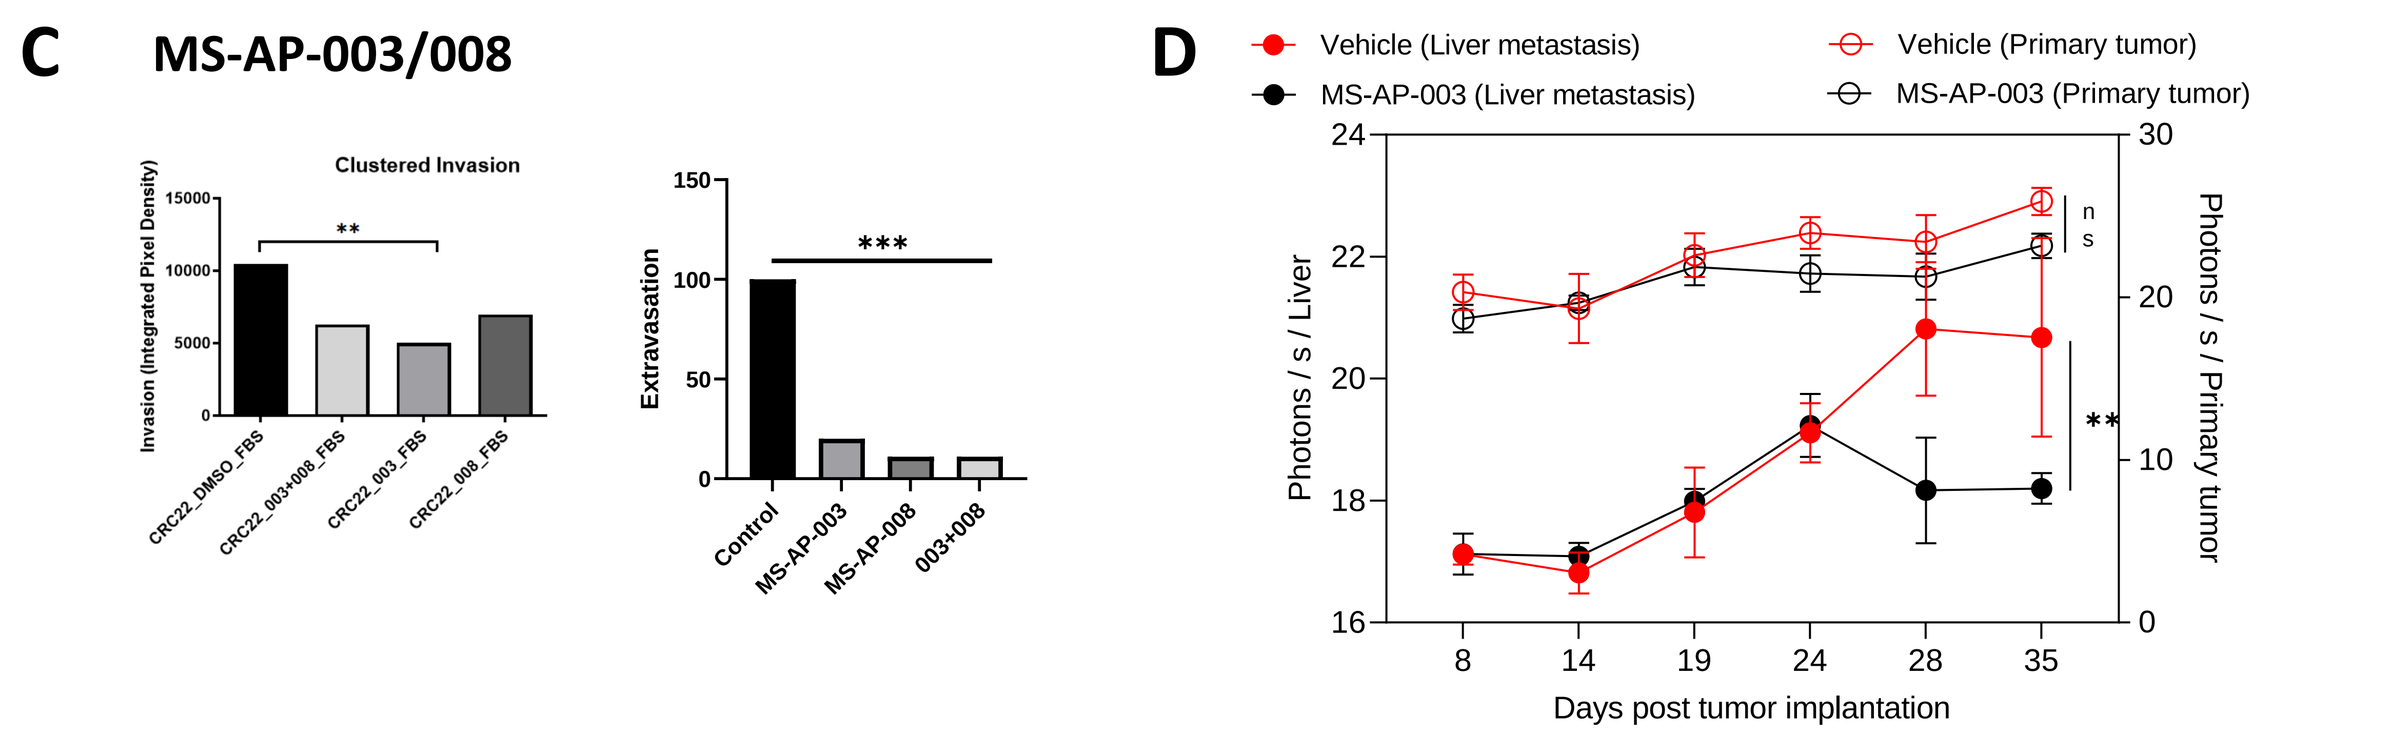
C
MS-AP-003/008
Clustered Invasion
Extravasation
D
Vehicle (Liver metastasis)
MS-AP-003 (Liver metastasis)
Vehicle (Primary tumor)
MS-AP-003 (Primary tumor)
Photons / s / Liver
Photons / s / Primary tumor
Days post tumor implantation

Fig.3: Effect of negative compound AP-006 on (E) patients and (F) in an animal model

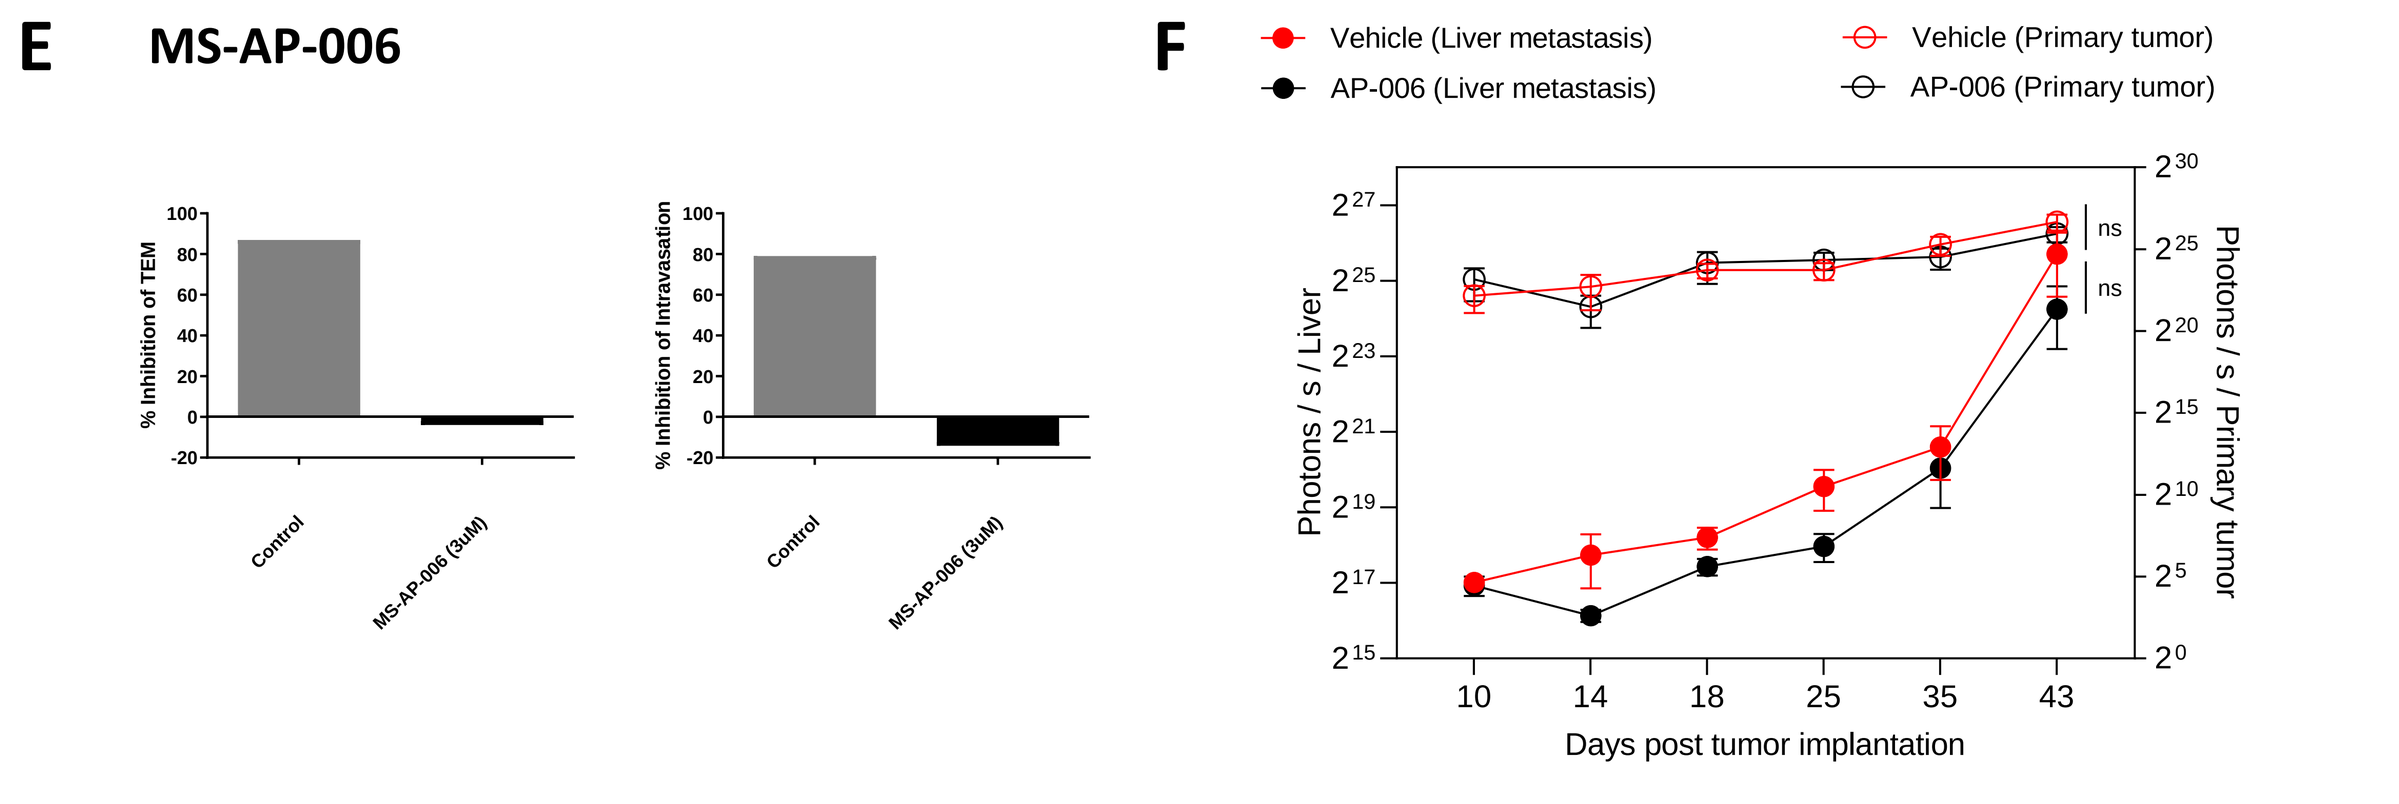
E
MS-AP-006
F
Vehicle (Liver metastasis)
AP-006 (Liver metastasis)
Vehicle (Primary tumor)
AP-006 (Primary tumor)
Photons / s / Liver
Photons / s / Primary tumor
Days post tumor implantation

Mestastop platforms identify anti-metastatic compounds, with in-vitro to in-vivo PoC

Results — Retrospective Clinical Study

Fig.4: (A) Study Design

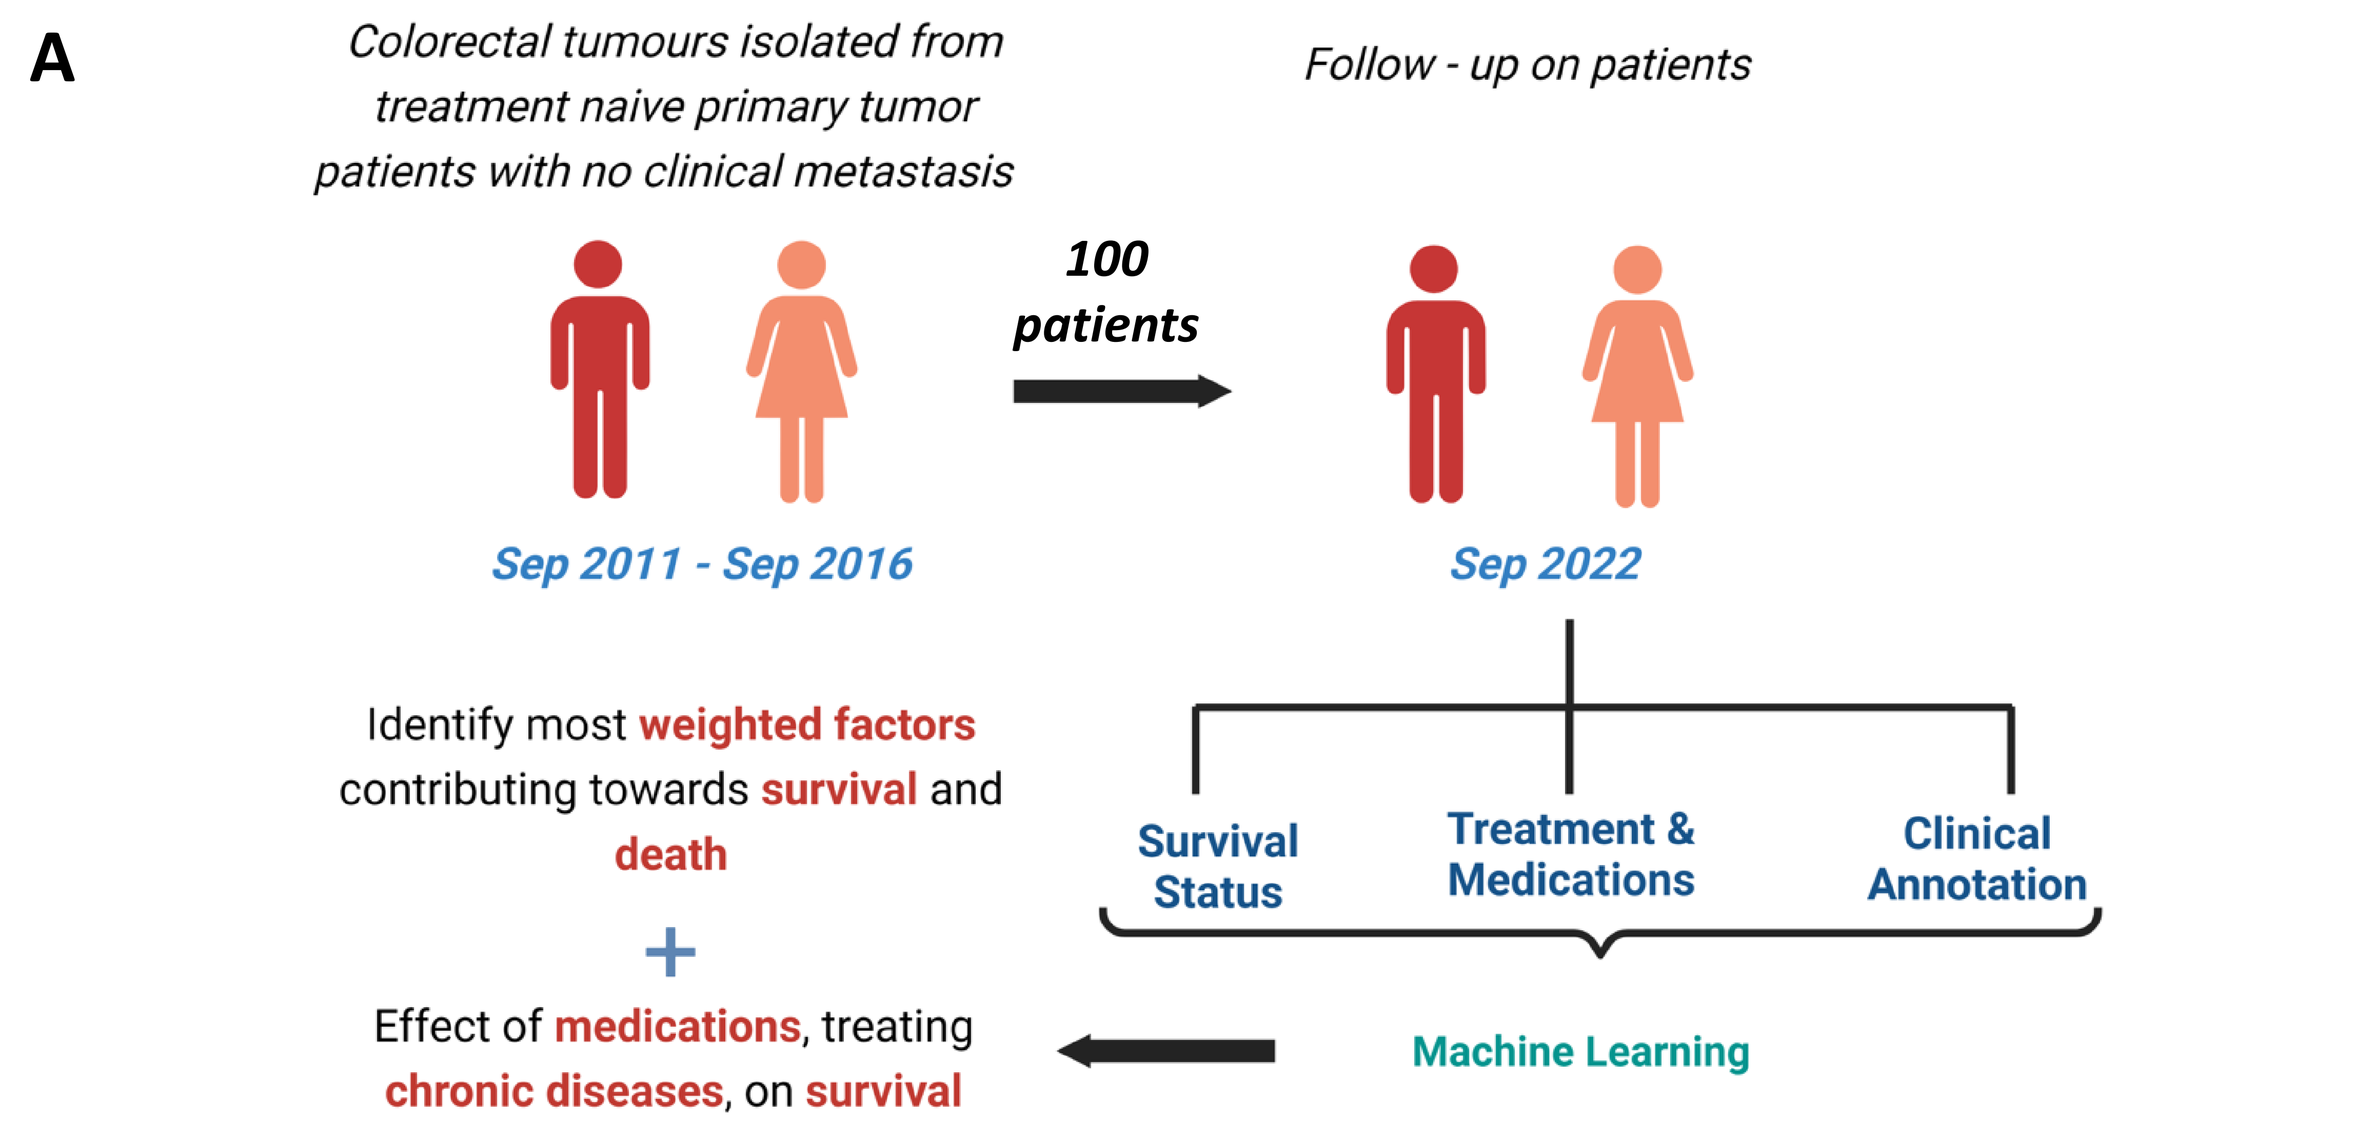
A
Colorectal tumours isolated from treatment naive primary tumor patients with no clinical metastasis
Follow - up on patients
100 patients
Sep 2011 - Sep 2016
Sep 2022
Identify most weighted factors contributing towards survival and death
Survival Status
Treatment & Medications
Clinical Annotation
Effect of medications, treating chronic diseases, on survival
Machine Learning

Fig.4: (B) Identification of weighted factors and (C) medicines with impact on survival

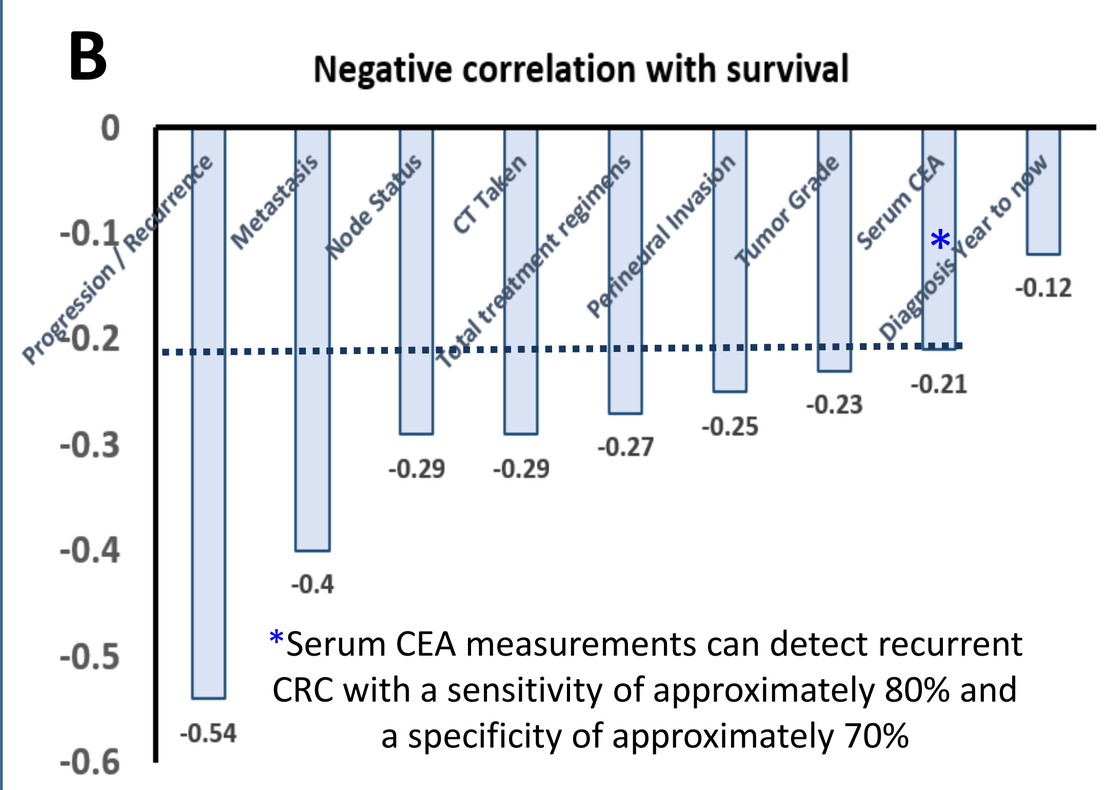
B
Negative correlation with survival
Progression / Recurrence
Metastasis
Node Status
CT Taken
Total treatment regimens
Perineural Invasion
Tumor Grade
Serum CEA
Diagnosis Year to now
*Serum CEA measurements can detect recurrent CRC with a sensitivity of approximately 80% and a specificity of approximately 70%

C

Medicine	Null Hypothesis (H0)	Rejects H0 (co-efficient < 0.05)	Co-efficient strength	p-value	Effect on Survival
A	The medications have a negative effect on the survivability of the subject	Yes	-0.33	0.03	Moderately positive
B		Yes	-0.24	0.01	Weakly positive
C		Yes	-0.03	0.046	Weakly positive
D		No	-0.15	0.55	more data required
E		Yes	-0.23	0.03	Weakly positive
F		Yes	-0.005	0.013	Weakly positive
G	The medications have a positive effect on the survivability of the subject	Yes	-0.19	0.03	Weakly negative
H		Yes	-0.31	0.003	Moderately negative
I		Yes	-0.33	0.03	Moderately negative
J		Yes	-0.2	0.01	Weakly negative

Retrospective study confirms effect of non-oncology drugs on survival

Results — Retrospective trial PoC

Fig.5: Effect of approved drugs in (A) retrospective study survival and (B) METSCAN®

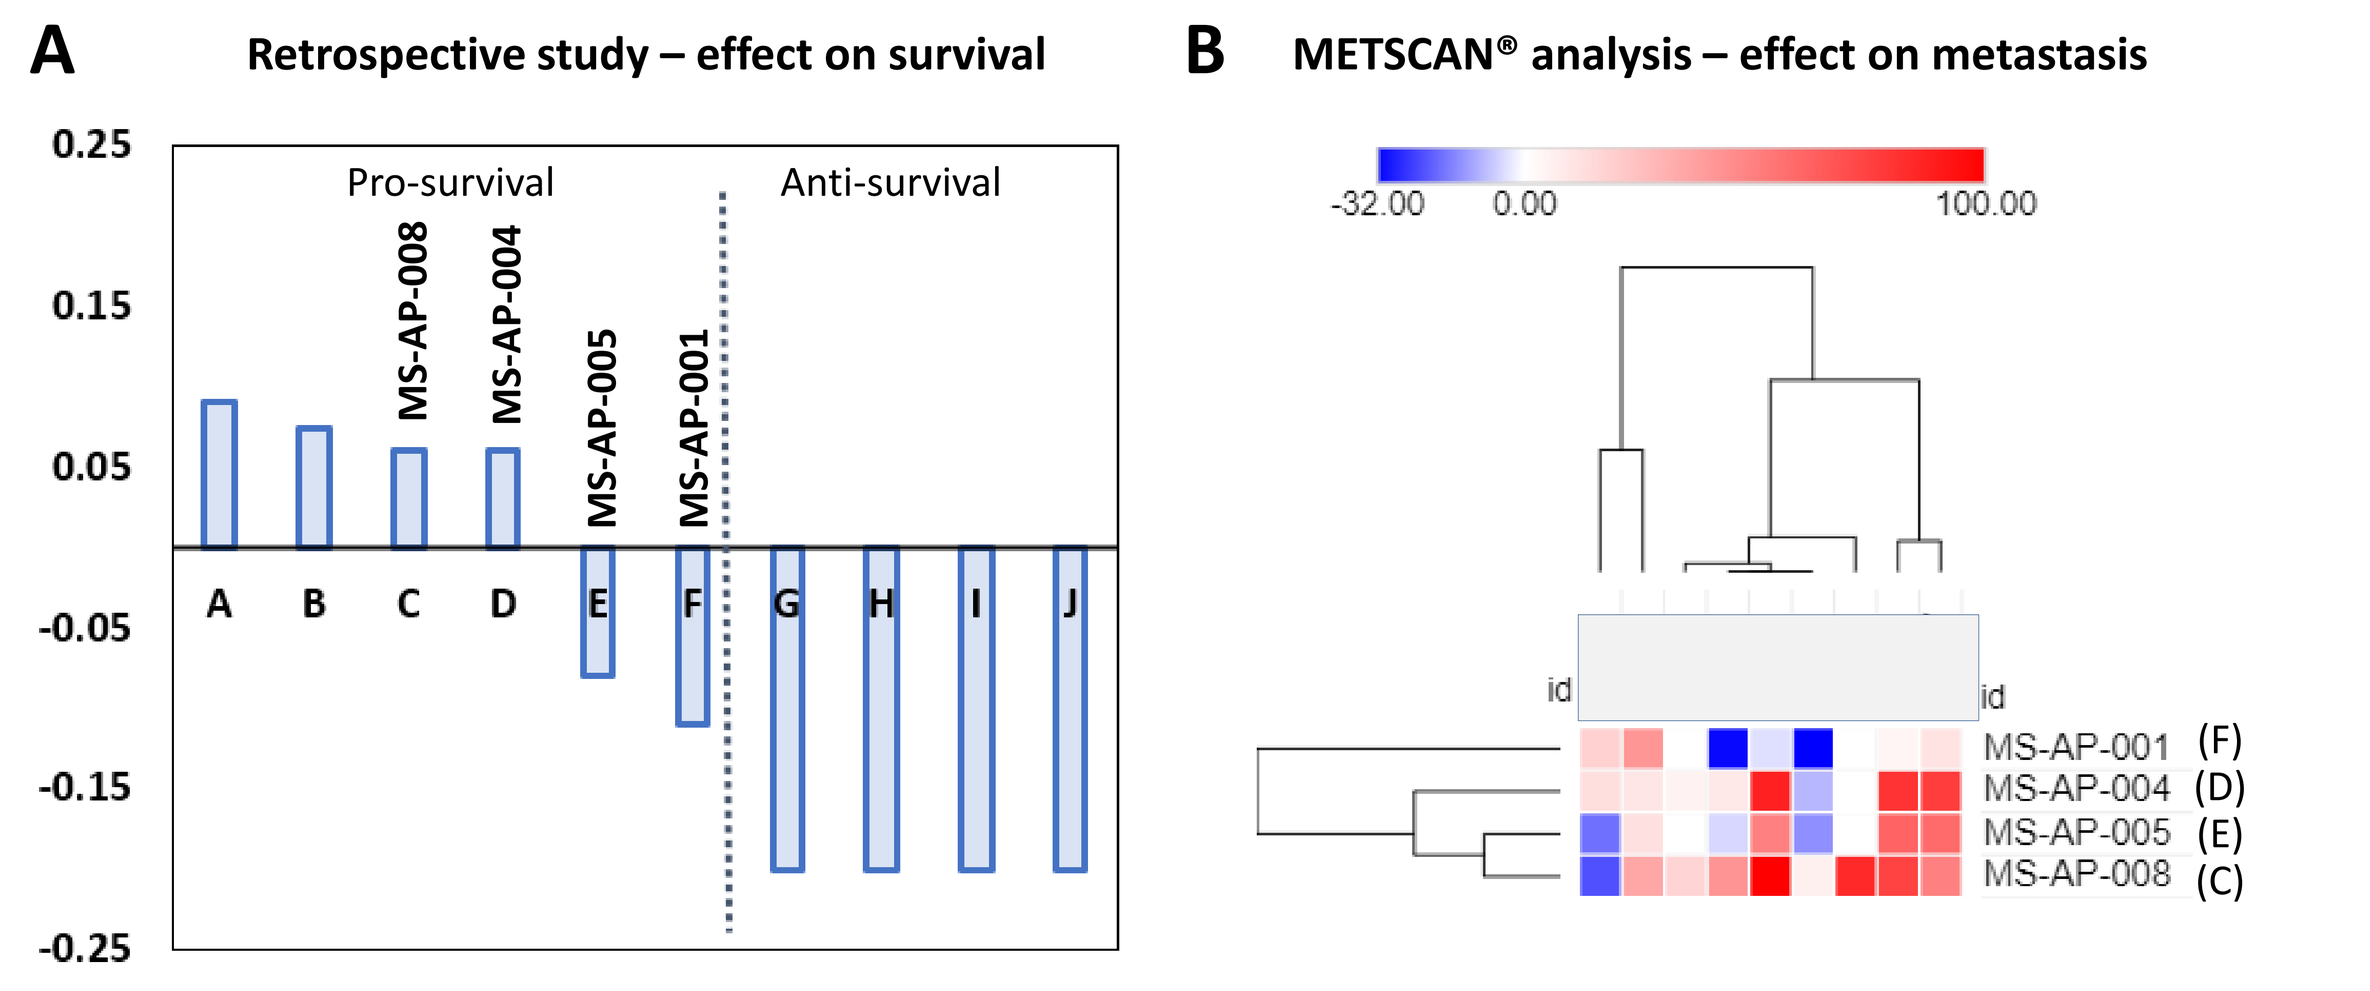
A
Retrospective study – effect on survival
Pro-survival
Anti-survival
MS-AP-008
MS-AP-004
MS-AP-005
MS-AP-001
B
METSCAN® analysis – effect on metastasis
MS-AP-001 (F)
MS-AP-004 (D)
MS-AP-005 (E)
MS-AP-008 (C)

All four drugs identified in patients, picked up by METSCAN®, inhibiting metastasis

Fig. 6: Analysis of the effect of approved drug A (MS-AP-031), identified by the retrospective study on the METSCAN® platform

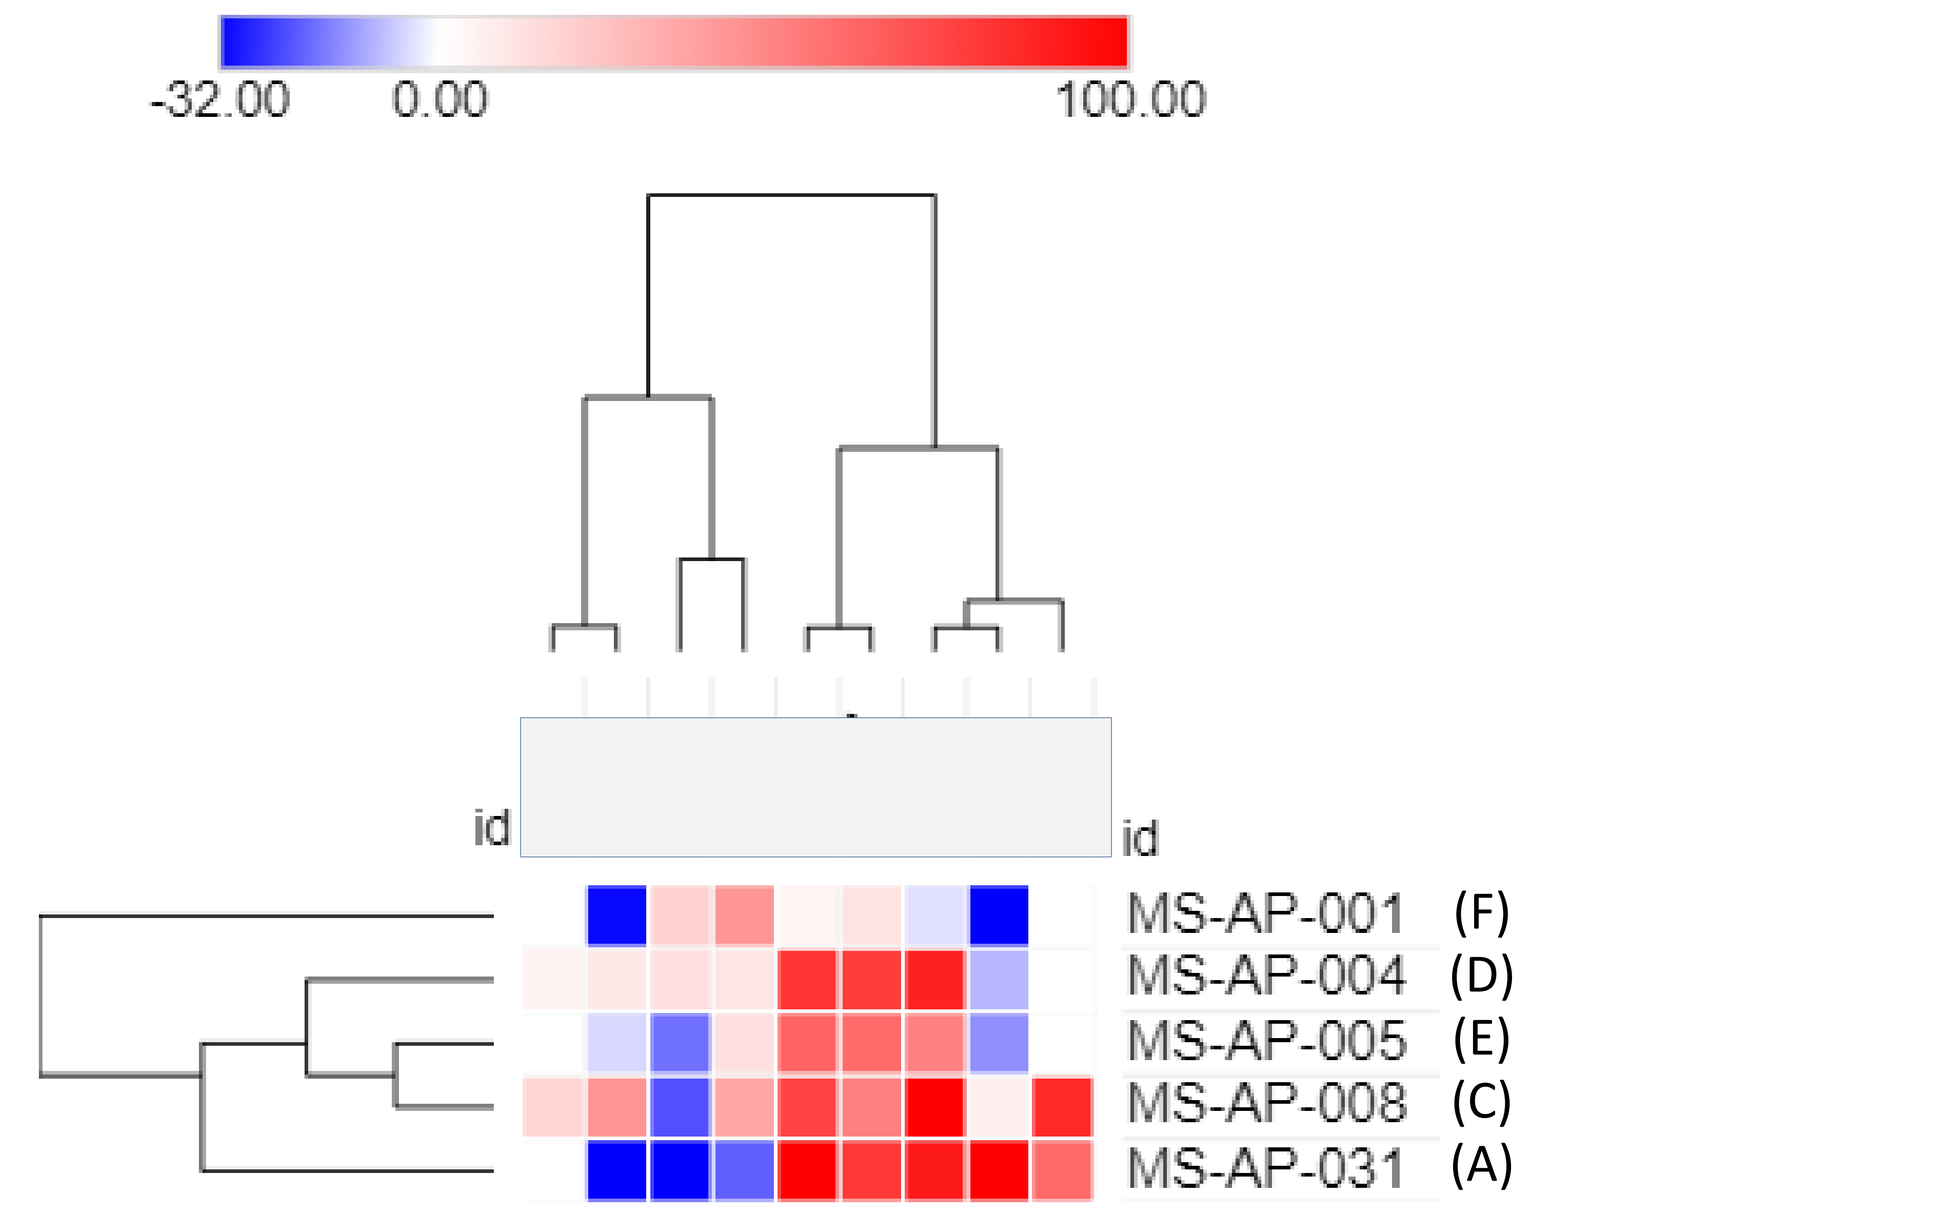
-32.00 0.00 100.00
MS-AP-001 (F)
MS-AP-004 (D)
MS-AP-005 (E)
MS-AP-008 (C)
MS-AP-031 (A)

METSCAN® rank ordering closely resembles the retrospective study survival data

Results — Cytotoxicity profiling

Fig.7: Inhibition of (A) metastatic steps not due to (B) cytotoxicity

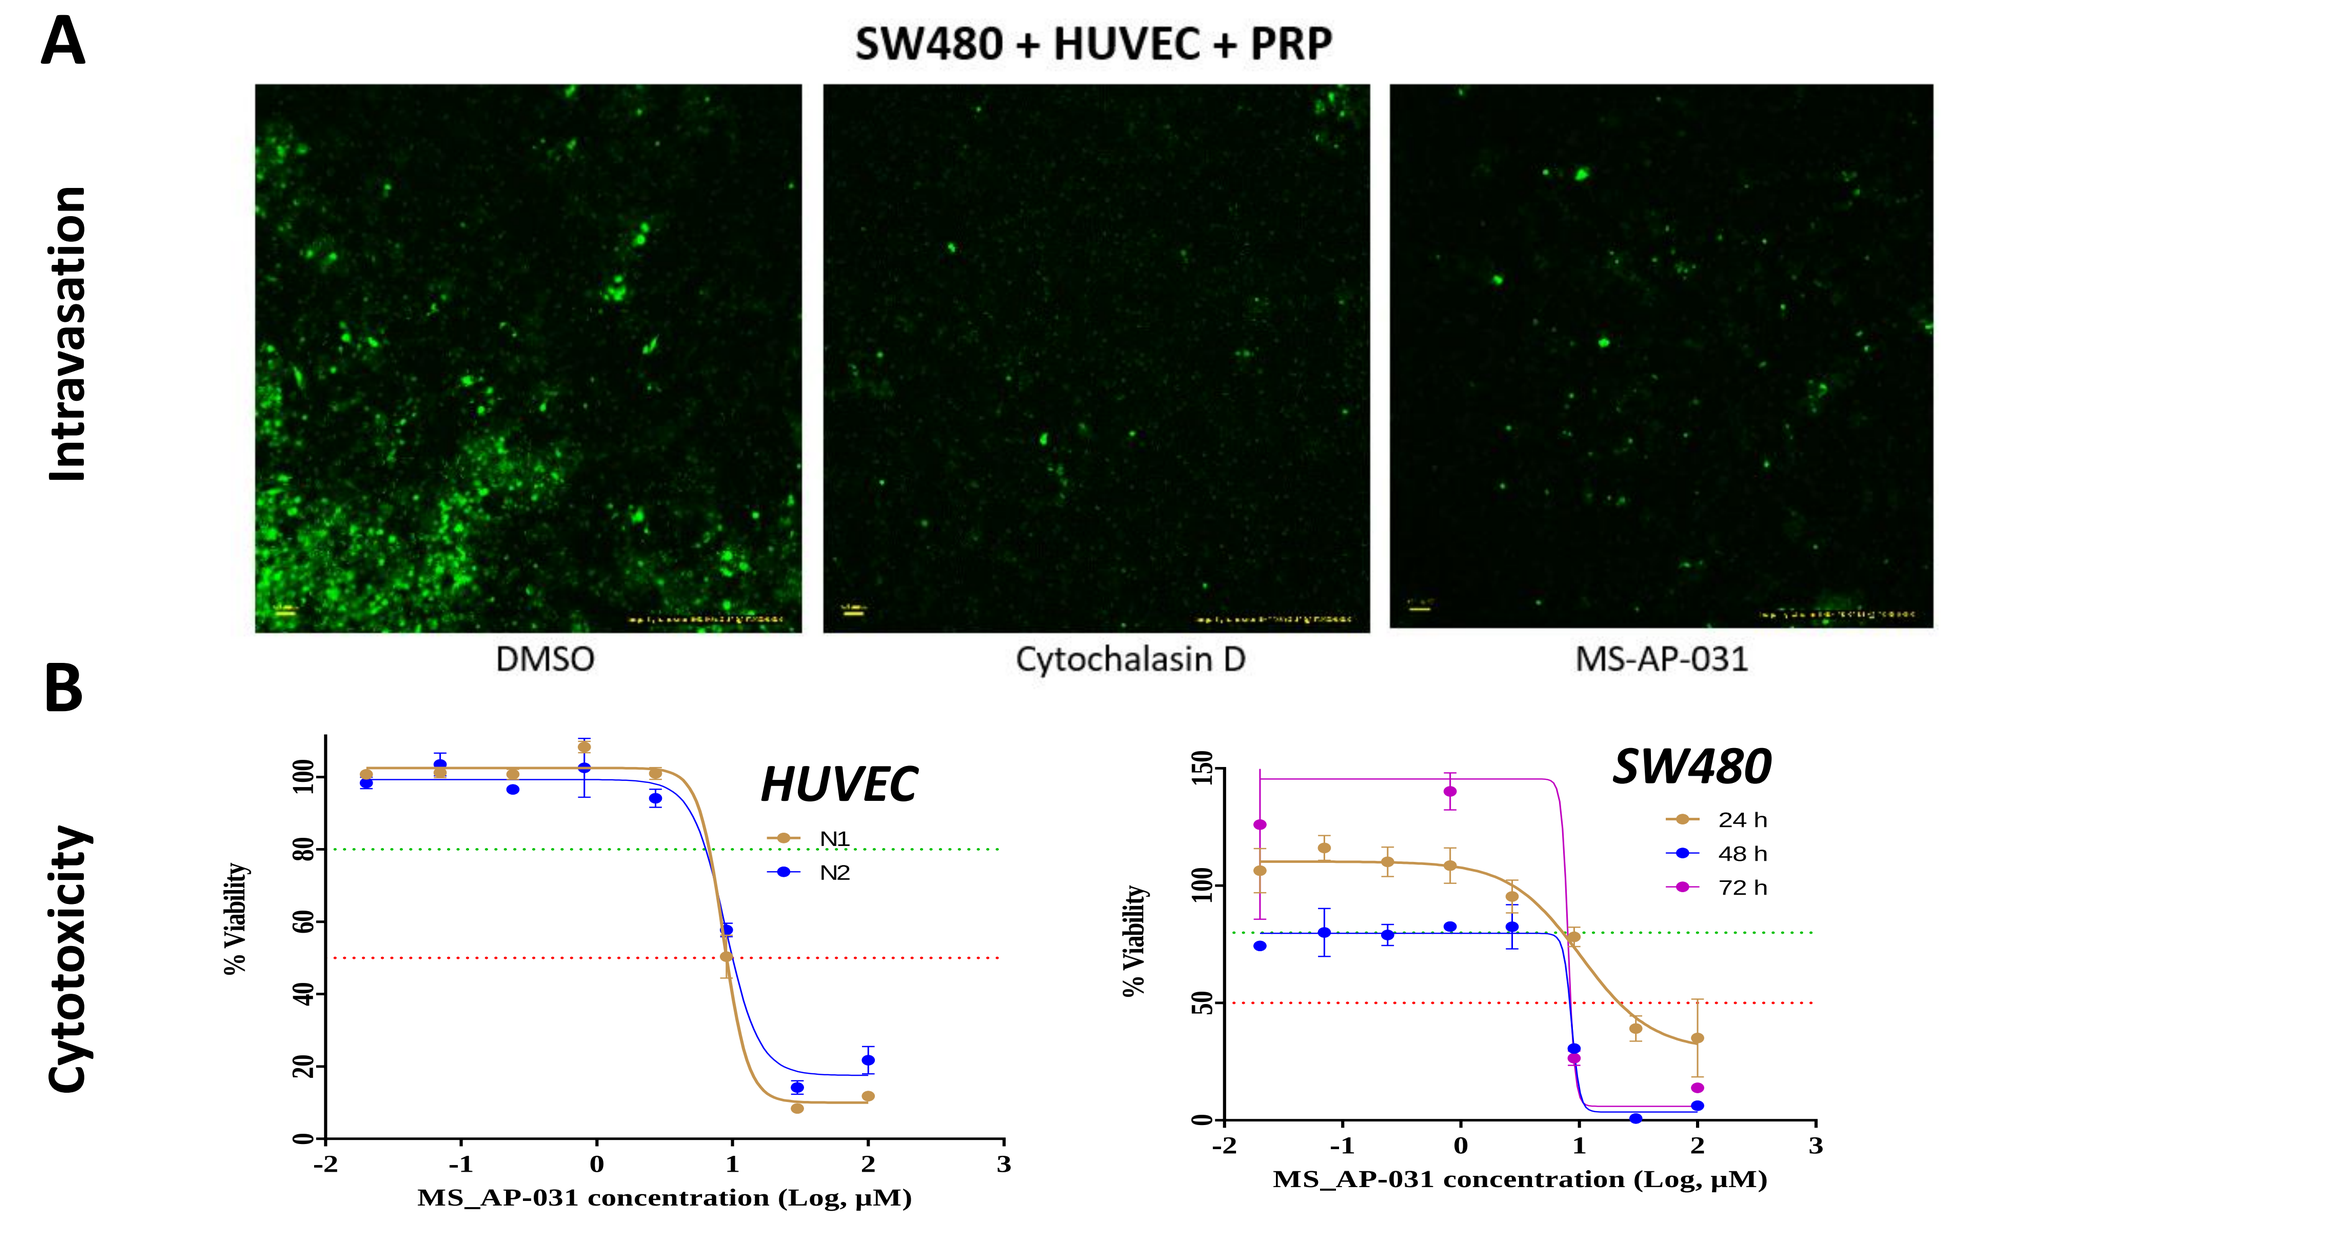
A
SW480 + HUVEC + PRP
Intravasation
DMSO
Cytochalasin D
MS-AP-031
B
Cytotoxicity
HUVEC
SW480
% Viability
MS_AP-031 concentration (Log, μM)

All METSCAN® assays performed at non-cytotoxic concentrations (cell lines & time)

Conclusion

1. METAssay® can successfully triage molecules and identify potent anti-metastasis compounds based on the weighted algorithm of patient-derived METSCAN®
2. The identified compounds translated in the in vivo animal model, METVivo®
3. Retrospective clinical trial of 100 CRC patients suggests that approved non-oncology drugs can impact the survival of primary cancer patients over a follow-up period of five years.
4. Four of these drugs were previously tested in the METSCAN® platform, and rank ordering from the platform almost matched with the retrospective clinical trial data (75%).
5. Further analysis of another compound, Drug A (MS-AP-031), increased the efficiency to 80%.

Next Steps

1. Currently running another retrospective study with 100 head and neck cancer patients
2. Planning for 2000 patient sample retrospective study in Europe and USA
3. Testing more compounds identified by the current study (both with positive and negative impact on survival) on the METSCAN® platform
4. FDA-Approved drug library testing on the METSCAN® platform
5. Identification of potential combinations
6. Fixed dose combination patent followed by clinical trials

References

1. Nature Reviews Clinical Oncology, 2019, 16, 185-204
2. J Clin Oncol., 2022 Dec 10; 40(35):4095-4106
3. Nature Reviews Clinical Oncology, 2022, 19, 486-492
4. Cancer Res 2021;81(13_Suppl):Abstract nr 2868 & Abstract nr 2841
5. EACR-MRS Conference on Seed and Soil: In Vivo Models of Metastasis, Jan 2022, Poster #24
6. MRS 19th Biennial Congress, Nov 2022, Abstract no. 1064653

Rajiv Gandhi Cancer Institute and Research Centre

https://mestastop.com/

Contact us: bd@mestastop.com

https://www.linkedin.com/company/mestastop-solutions/